Poster
PF1002

Prognostic value of circulating tumor DNA (ctDNA) detection by PhasED-Seq after axicabtagene ciloleucel therapy in relapsed/refractory large B-cell lymphoma

Brodie Miles[1], Daqin Mao[1], David M. Kurtz[2], Brian J. Sworder[2], Sierra Love Stowell[2], Hayley Warsinske[2], Saran Vardhanabhuti[1], Mike Mattie[1], Simone Filosto[1], Davide Bedognetti[1], and Rhine Shen[1]

[1]Kite, a Gilead company, Santa Monica, United States of America, [2]Foresight Diagnostics, Boulder, United States of America

BACKGROUND

- Axicabtagene ciloleucel (axi-cel) is an autologous chimeric antigen receptor (CAR) T-cell therapy approved for adults with relapsed/refractory large B-cell lymphoma (LBCL) based on significant clinical benefit demonstrated in second line (2L) of therapy based on Zuma-7 results[1]
 - Despite strong response rates to axi-cel therapy, many subjects in Zuma-7 eventually experience disease progression (4 year estimated event-free survival rates of 39% in axi-cel and 17% in standard of care (SOC) patients)[2]
- Monitoring of ctDNA levels in blood using the ultrasensitive phased variant enrichment and detection sequencing (PhasED-Seq) technology has previously been used to assess minimal residual disease (MRD) in LBCL
 - PV detection at interim and EOT were more predictive than PET/CT in lymphoma patients receiving first line SOC therapy[3]
 - PV detection at EOT in 1L SOC therapy showed very high prognostic value compared to PET/CT and was able to adjudicate patients with a EOT PET/CT CR who relapsed[4]

OBJECTIVES

- Here, we explored the value of ctDNA detection by PhasED-Seq to predict outcomes of patients receiving axi-cel in r/r LBCL
- We investigated the association of event free and progression free survival (Zuma-7 primary and secondary endpoints) with MRD at early timepoints post CAR T cell infusion with the aim of determining a landmark for MRD analysis in CAR T therapy
- At these early timepoints, we investigated the concordance of MRD results with corresponding PET/CT imaging results

METHODS

- Patients treated with either axi-cel or standard of care (SOC) with evaluable pre-infusion tumor and post-infusion peripheral tracking plasma ctDNA (n=44) were analyzed by the Foresight CLARITY assay, powered by PhasED-Seq. Tumor informed variants were identified in biopsy tissue obtained prior to treatment and longitudinally monitored in plasma at approximately 50, 100, and 150 days post-randomization. On average, 0.8 mL (range 0.2-2.1mL) of plasma per timepoint was available for phased variant (PV) tracking. Positive predictive value (PPV; MRD+ pts who relapsed or were nonresponders/total MRD+ pts ×100) and negative predictive value (NPV; MRD- pts in ongoing responders/total MRD- pts ×100) were assessed at days 50 and 150.

RESULTS

Figure 1. Survival stratified by MRD status at day 50 post randomization

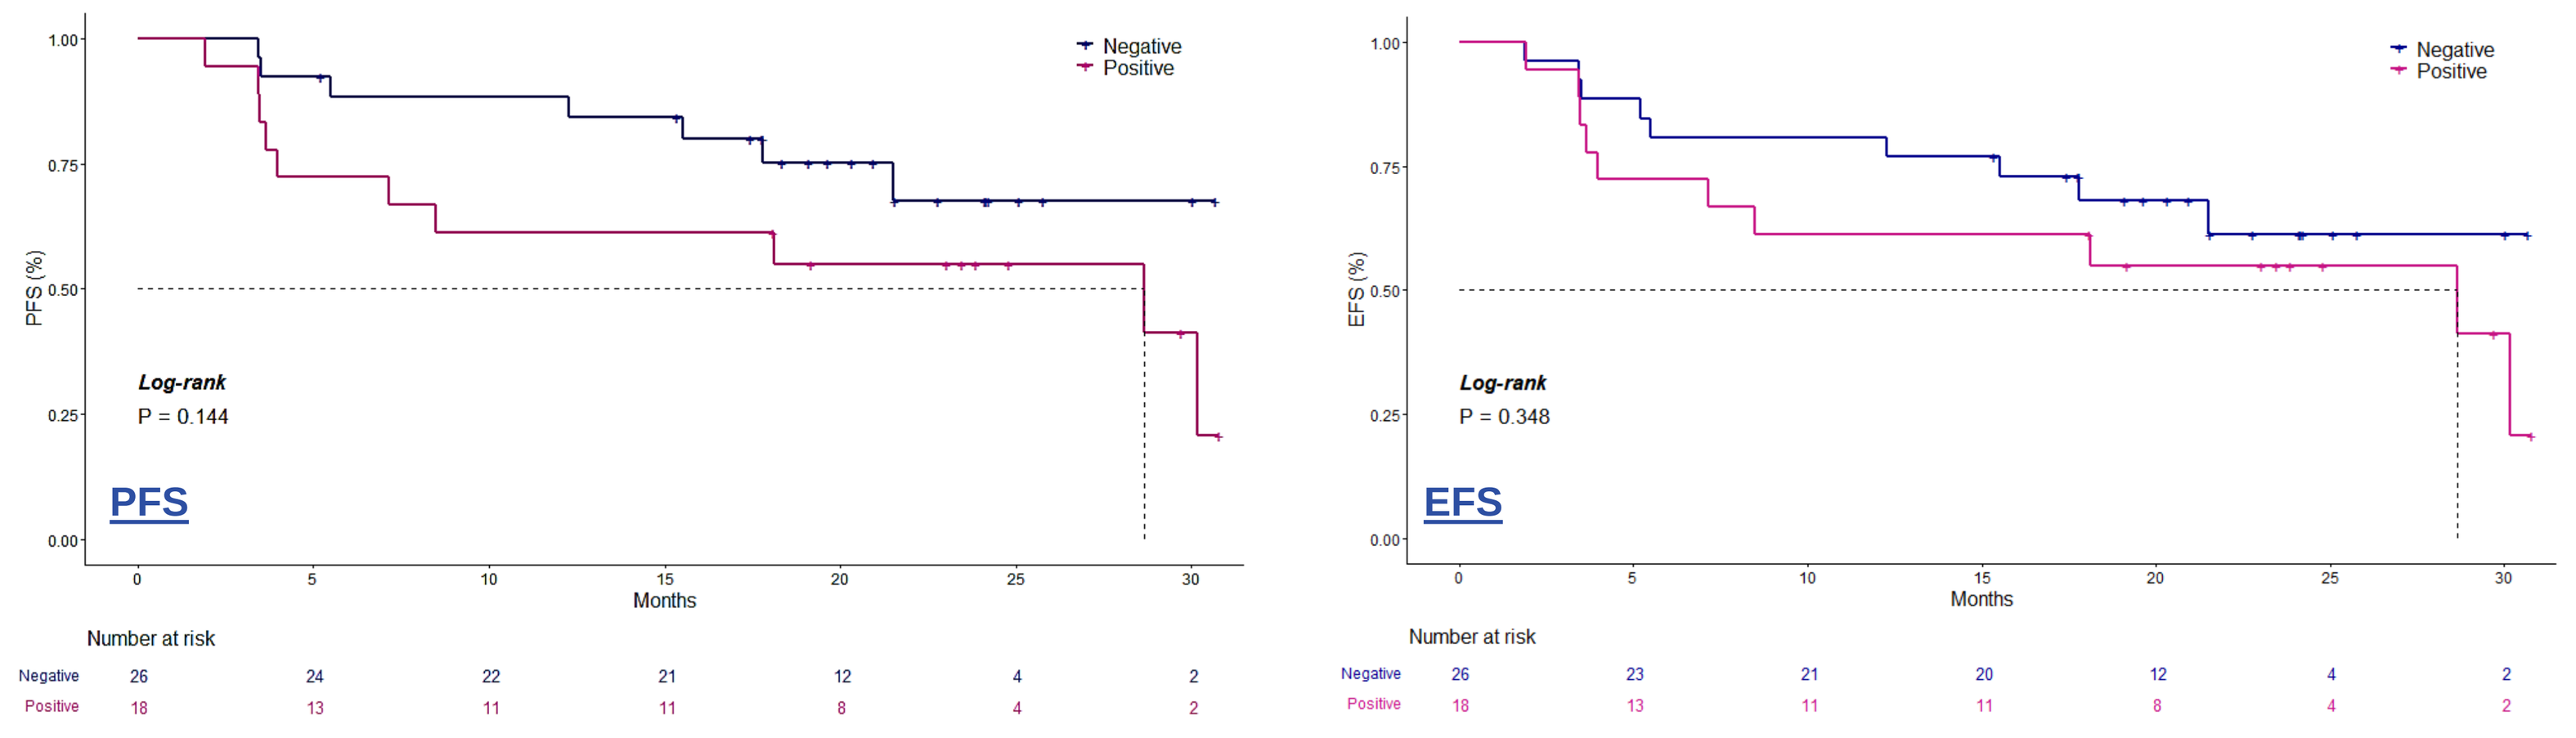
Negative
Positive
Log-rank
P = 0.144
PFS
Months
Number at risk
Negative 26 24 22 21 12 4 2
Positive 18 13 11 11 8 4 2
Log-rank
P = 0.348
EFS
Number at risk
Negative 26 23 21 20 12 4 2
Positive 18 13 11 11 8 4 2

- This analysis consists of grouped axicel (n=39) and SOC (n=5) patients
- Day 50 post-randomization is approximately 14-28 days post-axicel infusion and interim to SOC end of treatment (between cycles 3, 4)
- Significant differences between MRD negative and MRD positive patients were not observed based on MRD result at day 50

Figure 2. Association of survival based on MRD status at day 150 post randomization

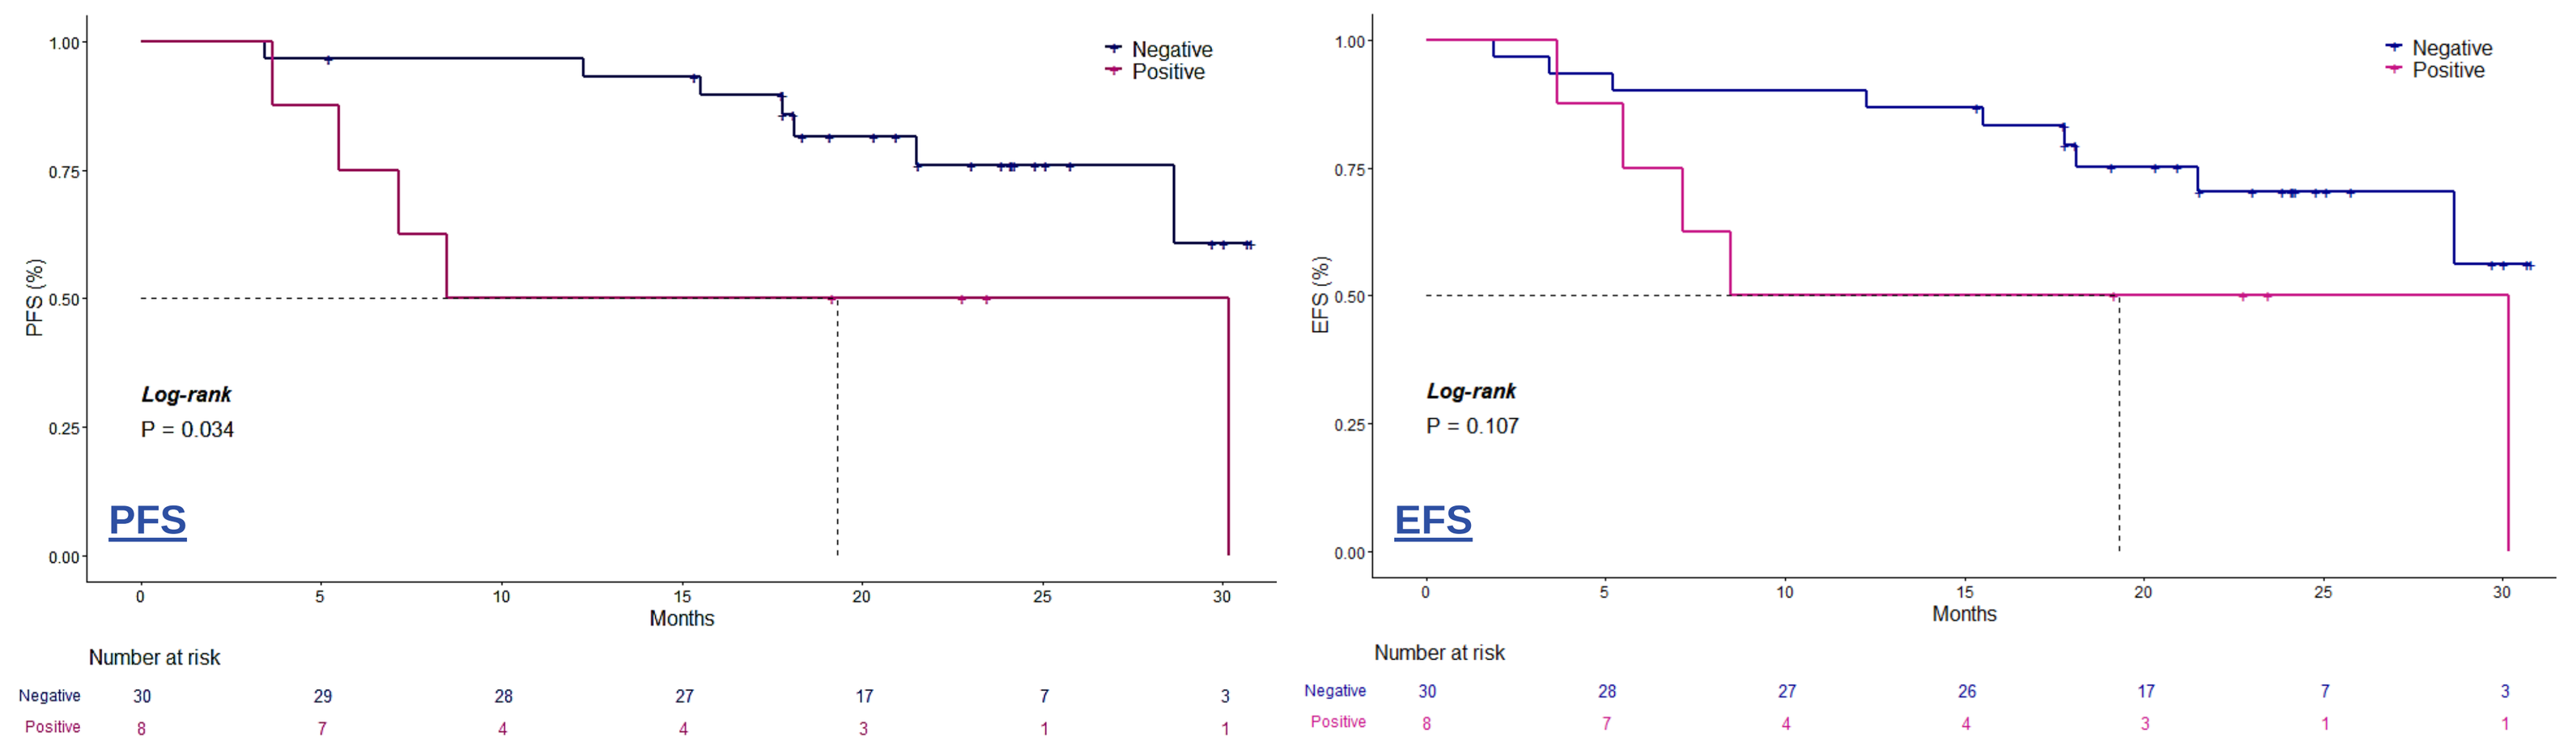
Negative
Positive
Log-rank
P = 0.034
PFS
Months
Number at risk
Negative 30 29 28 27 17 7 3
Positive 8 7 4 4 3 1 1
Log-rank
P = 0.107
EFS
Number at risk
Negative 30 28 27 26 17 7 3
Positive 8 7 4 4 3 1 1

- This analysis consists of grouped axicel (n=35) and SOC (n=3) patients
- Day 150 post-randomization is approximately 2-3 months post-axicel infusion and approximately 1 month post-SOC end of treatment
- Patients achieving MRD negativity by day 150 had significantly improved PFS and trended towards improved EFS compared to patients who remained MRD positive

Table 1. MRD positive and negative predictive values

MRD status	Ongoing Response	Relapsed	Non-responder	Predictive value
Timepoint, result	**n (%)**	**n (%)**	**n (%)**	**n (%)**
Day 50	26	15	2	
MRD negative	18 (69)	6 (40)	1 (50)	NPV[1]: 18/25 (72)
MRD positive	8 (31)	9 (60)	1 (50)	PPV[2]: 10/18 (56)
Day 150	25	12	0	
MRD negative	22 (88)	7 (58)	n/a	NPV[1]: 22/29 (76)
MRD positive	3 (12)	5 (42)	n/a	PPV[2]: 5/8 (63)

- The negative predictive value (all patients in ongoing response with MRD negative divided by all MRD negative patients) improved from day 50 to day 150 while the positive predictive value (all relapsed and non-responding patients with MRD positivity divided by all MRD positive patients) improved from day 50 to day 150
- [1]The negative predictive value was likely underestimated due to low plasma volumes (<1 mL), which yielded limited cfDNA and may have reduced assay sensitivity in MRD-negative cases.
- [2]The observed PPV reflects relapse status at last follow-up and does not account for censoring or patients with subsequent MRD clearance (e.g., positive at day 50 and negative at day 150). Most MRD-positive patients who subsequently maintained MRD positivity ultimately recurred. suggesting the PPV with adequate follow-up may be higher

Figure 3. Improved survival by overall undectable MRD status

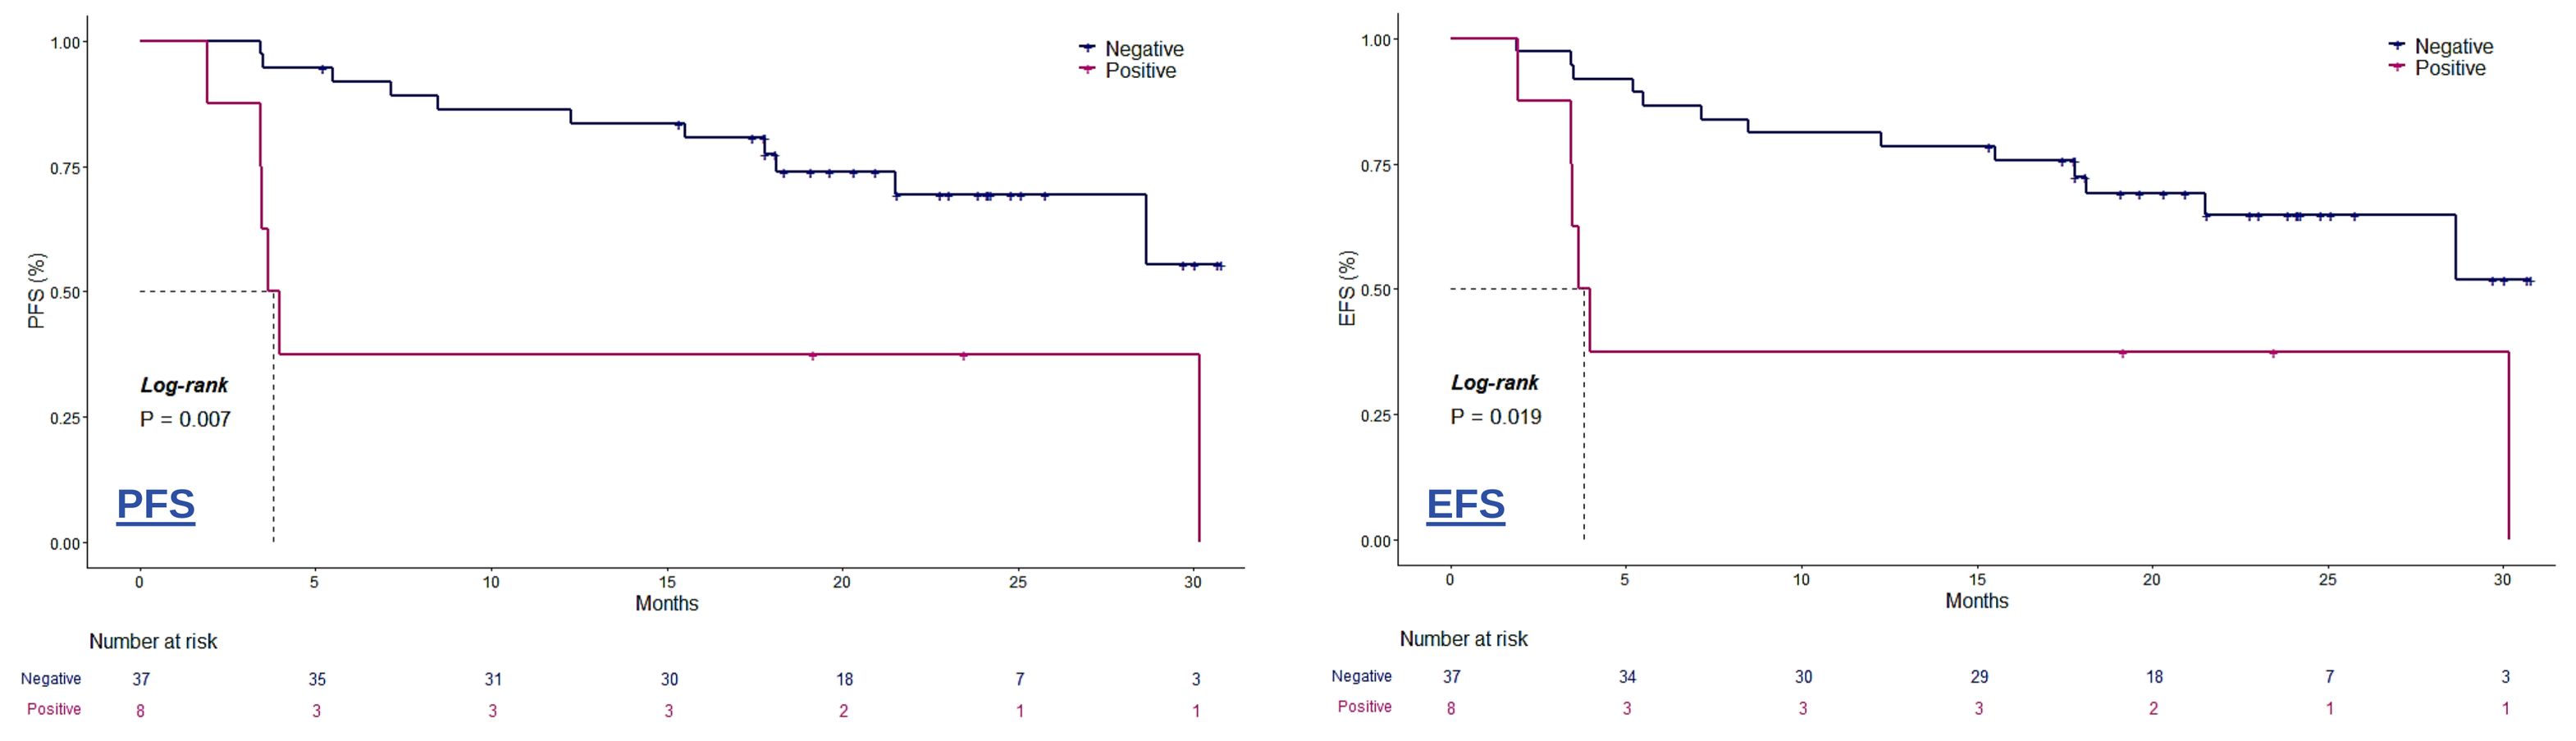
Negative
Positive
Log-rank
P = 0.007
PFS
Months
Number at risk
Negative 37 35 31 30 18 7 3
Positive 8 3 3 3 2 1 1
Log-rank
P = 0.019
EFS
Number at risk
Negative 37 34 30 29 18 7 3
Positive 8 3 3 3 2 1 1

- This analysis consists of grouped axicel (n=40) and SOC (n=5) patients
- Patients who achieve MRD negativity at any timepoint post treatment (in day 50, day 100, or day 150 post-randomization tracking samples) have a significantly improved duration of both PFS and EFS compared to those who remain MRD positive

Figure 4. Association of survival based on MRD result at the last sample tested

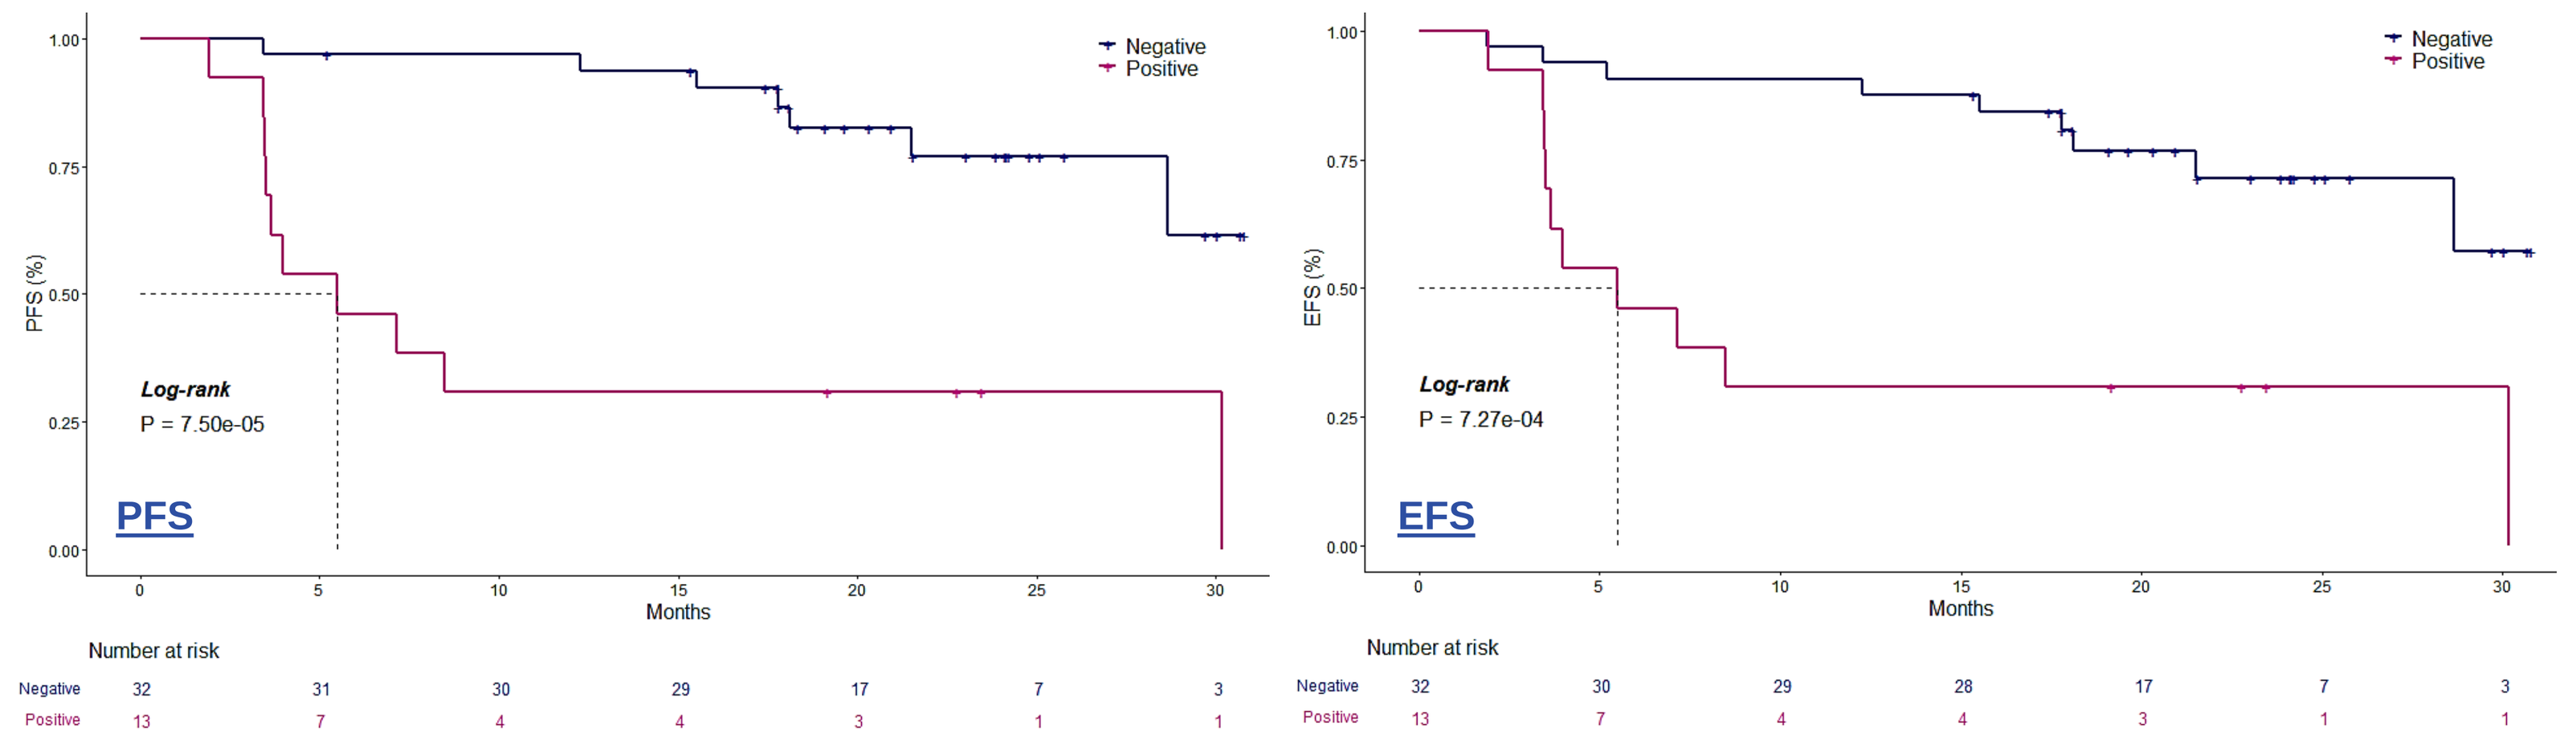
Negative
Positive
Log-rank
P = 7.50e-05
PFS
Months
Number at risk
Negative 32 31 30 29 17 7 3
Positive 13 7 4 4 3 1 1
Log-rank
P = 7.27e-04
EFS
Number at risk
Negative 32 30 29 28 17 7 3
Positive 13 7 4 4 3 1 1

- This analysis consists of grouped axicel (n=40) and SOC (n=5) patients
- Patients who achieve an MRD negative result at their last sample tested (day 50, day 100, and day 150 post-randomization tracking samples) have a significantly improved duration of both PFS and EFS compared to those who remain MRD positive

Table 2. MRD shows CR concordance and detects high non-CR conversion rate

Time of Assessment	MRD negative	MRD positive	MRD concordance with CR rate
PET/CT result	**(n)**	**(n)**	**(X%)**
Day 50			
CR	21	9#	70
Non-CR	5^	9	64
Day 150			
CR	27	7*	79
Non-CR	3X	1	25

- Patients were divided based on those who achieved a PET/CR CR and those who did not achieve a CR at both day 50 and day 150
- The MRD testing result at the corresponding timepoint was then assessed to determine the concordance to CR and non-CR

Day 50

- #Of the 9 patients achieving CR at day 50 with MRD positivity, 5 subsequently relapsed and 2 subsequently achieved MRD negativity
- ^Of the 5 patients without a CR at day 50 with MRD negativity, 1 patient had stable disease and became MRD positive at subsequent testing, while 4 patients subsequently achieved CR

Day 150

- *Of the 7 patients achieving CR at day 150 with MRD positivity, 4 patients subsequently relapsed
- XOf the 3 patients without a CR by day 150 with MRD negativity, 2 patients subsequently achieved CR

CONCLUSIONS

- Although a limited number of subjects, small plasma volumes, and low quantities of tumor DNA for assay personalization were available, PhasED-seq MRD provided prognostic information at day 50, with improved performance observed by day 150. The negative predictive value may have been underestimated due to low plasma volumes (~1 mL), which limited cfDNA input and potentially reduced assay sensitivity
- MRD negativity at later timepoints was more strongly associated with improved PFS than early MRD negativity, highlighting the potential value of serial assessment
- Concordance between MRD negativity and CR by PET/CT improved over time, increasing from day 50 to day 150
- This analysis highlights the prognostic value of MRD detection by ultrasensitive measures, while further supporting a role for longitudinal monitoring to assess the unique timing of MRD clearance during CD19 CAR T cell therapy
- Taken together, these findings demonstrate the prognostic utility of ultra-sensitive MRD detection early in CAR T therapy and support further investigation of MRD as a surrogate endpoint in LBCL

REFERENCES

1. Locke FL, et al. *N Eng J Med*. 2022;386:640-654.
2. Westin JR, et al. *N Eng J Med*. 2023;389:148-157.
3. Sworder BJ. et al. Oral abstract, ASH 2023
4. Roschewski, M. et al. Oral abstract, ASH 2023

ACKNOWLEDGMENTS

- We thank the patients, families, friends, and caregivers associated with this study
- We also thank the study investigators, coordinators, and health care staff at each study site
- This study was funded by Kite, a Gilead Company

DISCLOSURES

BM: employment with Kite, a Gilead company; and stock or other ownership in Gilead Sciences; **DM**: employment with Kite, a Gilead company; and stock or other ownership in Gilead Sciences; **DMK**: Employment with Foresight Diagnostics; and stock or other ownership in Foresight Diagnostics; **BJS**: Employment with Foresight Diagnostics; and stock or other ownership in Foresight Diagnostics; **SLS**: Employment with Foresight Diagnostics; and stock or other ownership in Foresight Diagnostics; **HW**: Employment with Foresight Diagnostics; and stock or other ownership in Foresight Diagnostics; **SV**: employment with Kite, a Gilead company; and stock or other ownership in Gilead Sciences; **MM**:employment with Kite, a Gilead company; and stock or other ownership in Gilead Sciences; **SF**:employment with Kite, a Gilead company; and stock or other ownership in Gilead Sciences; **DB**:employment with Kite, a Gilead company; and stock or other ownership in Gilead Sciences; **RS**:employment with Kite, a Gilead company; and stock or other ownership in Gilead Sciences

Presented at the 2025 European Hematology Association Annual Congress